5 August 2026 Sydney, Australia

CSANZ26 Conference Poster

Nyrada Inc (ASX:NYR), a clinical-stage biotechnology company focused on developing Transient Receptor Potential Canonical (TRPC) ion channel inhibitors to treat a range of medical conditions, is pleased to provide a copy of a conference poster to be presented at the 74th Annual Scientific Meeting of the Cardiac Society of Australia and New Zealand (CSANZ26).

Nyrada's Director of Research and Development, Dr Jasneet Parmar, will be presenting at CSANZ 2026 to be held in Sydney from 6 to 9 August 2026. The poster presents preclinical data showing that Xolatryp® provided significant cardioprotection in two studies using a rat model of myocardial ischemia-reperfusion injury. The data from these studies show Xolatryp preserved myocardial structure and function while reducing ventricular arrhythmias following acute myocardial ischemia. Xolatryp is a small-molecule inhibitor of TRPC3/6/7 channels designed to limit excessive calcium entry related to multiple disease pathologies.

A Phase I clinical trial to assess the safety, tolerability, and pharmacokinetics has been successfully completed and a Phase IIa clinical trial has commenced to assess the safety and preliminary efficacy of Xolatryp in reducing cardiac reperfusion injury in patients with ST-Elevation Myocardial Infarction (STEMI) undergoing PCI.

Page 1

Nyrada Inc. Level 22/23, Salesforce Tower, 180 George Street, Sydney NSW 2000 ARBN 625 401 818

Key Links:

Latest Corporate Presentation - https://bit.ly/4v0QgXE

Foundation Studies

- GLP study results – https://bit.ly/4d8VYkX
- Phase I trial results – https://bit.ly/3Nt0GzH

Cardiac Studies

- Phase IIa PROTECT-MI website - https://www.protect-mi.com
- Phase IIa study fact sheet – https://bit.ly/4bcDlKj
- Preclinical cardioprotection study 1a - https://bit.ly/4sigwMZ
- Preclinical cardioprotection study 1b - https://bit.ly/4rmOsXn
- Preclinical cardioprotection study 2 - https://bit.ly/40jHpUg

Oncology Studies

- Preclinical anti-tumour study - https://bit.ly/49rWOa0
- Preclinical cardioprotection pilot study - https://bit.ly/3QS1VKm

Stroke Study

- Preclinical stroke study - https://bit.ly/4sygHmH

TBI Study

- Preclinical TBI study - https://bit.ly/40fhrRT

Page 2

Nyrada Inc. Level 22/23, Salesforce Tower, 180 George Street, Sydney NSW 2000 ARBN 625 401 818

About Nyrada Inc.

Nyrada Inc. is a clinical-stage biotechnology company focused on the discovery and development of innovative small-molecule therapies, specifically targeting Transient Receptor Potential Canonical (TRPC) ion channels. The company's lead candidate, Xolatryp®, has shown efficacy in preclinical cardioprotection, neuroprotection, and oncology models and has completed a first-in-human Phase I clinical trial. A Phase IIa clinical trial has commenced to assess the safety and preliminary efficacy of Xolatryp in reducing cardiac reperfusion injury in patients with ST-Elevation Myocardial Infarction (STEMI) undergoing PCI. Nyrada Inc. (ARBN 625 401 818) is incorporated in Delaware, US, with limited liability for its stockholders.

www.nyrada.com

Authorised by James Bonnar, Managing Director and CEO, on behalf of the Board.

Investor & Media Enquiries:
Dimitri Burshtein
T: 0491 789 391
E: info@nyrada.com

Company Secretary:
David Franks
T: 02 8072 1400
E: David.Franks@automicgroup.com.au

Forward-Looking Statements

This announcement may contain forward-looking statements. You can identify these statements by the fact they use words such as "aim", "anticipate", "assume", "believe", "continue", "could", "estimate", "expect", "intend", "may", "plan", "predict", "project", "plan", "should", "target", "will" or "would" or the negative of such terms or other similar expressions. Forward-looking statements are based on estimates, projections, and assumptions made by Nyrada about circumstances and events that have not yet taken place. Although Nyrada believes the forward-looking statements to be reasonable, they are not certain. Forward-looking statements involve known and unknown risks, uncertainties and other factors that are in some cases beyond the Company's control (including but not limited to the COVID-19 pandemic) that could cause the actual results, performance, or achievements to differ materially from those expressed or implied by the forward-looking statement.

Page 3

Nyrada Inc. Level 22/23, Salesforce Tower, 180 George Street, Sydney NSW 2000 ARBN 625 401 818

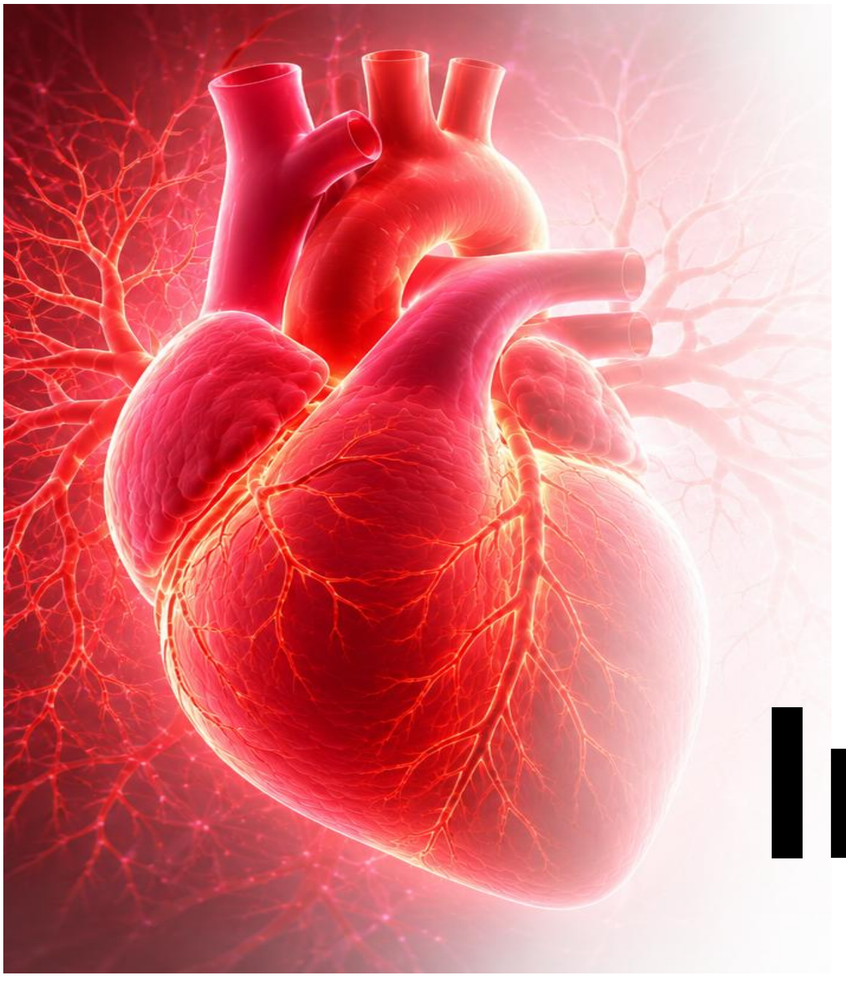

Xolatryp® - a Gαq-Coupled TRPC Channel Antagonist Attenuates Myocardial Reperfusion Injury and Malignant Arrhythmias in Rat MI Models

B. Evison[1], **J. Parmar[1,2]**, A. Suchowerska[1,2], P. Coghlan[1], G.D. Housley[2], J. Bonnar[1]
[1]Nyrada Inc., Sydney, NSW, Australia
[2]Translational Neuroscience Facility & Dept. Physiology, School of Biomedical Sciences, UNSW Sydney, Sydney, NSW, Australia

Background

- Primary percutaneous coronary intervention (PCI) restores coronary blood flow following ST-elevation myocardial infarction (STEMI), but reperfusion itself initiates secondary myocardial injury that contributes to infarct expansion, ventricular dysfunction and malignant arrhythmias.
- Pathological activation of Gαq-coupled receptor signalling during reperfusion stimulates TRPC3/6/7 channels, promoting excessive intracellular Ca^{2+} influx, mitochondrial dysfunction, oxidative stress and cardiomyocyte death.
- Xolatryp® is a potent, selective antagonist of TRPC3, TRPC6 and TRPC7 that targets this upstream calcium signalling pathway to preserve myocardial viability while maintaining physiological cardiac function.
- This study evaluated whether pharmacological inhibition of TRPC3/6/7 with Xolatryp reduces myocardial reperfusion injury and improves structural, functional and electrophysiological outcomes in rat models of acute myocardial infarction

TRPC ion channel signalling pathway

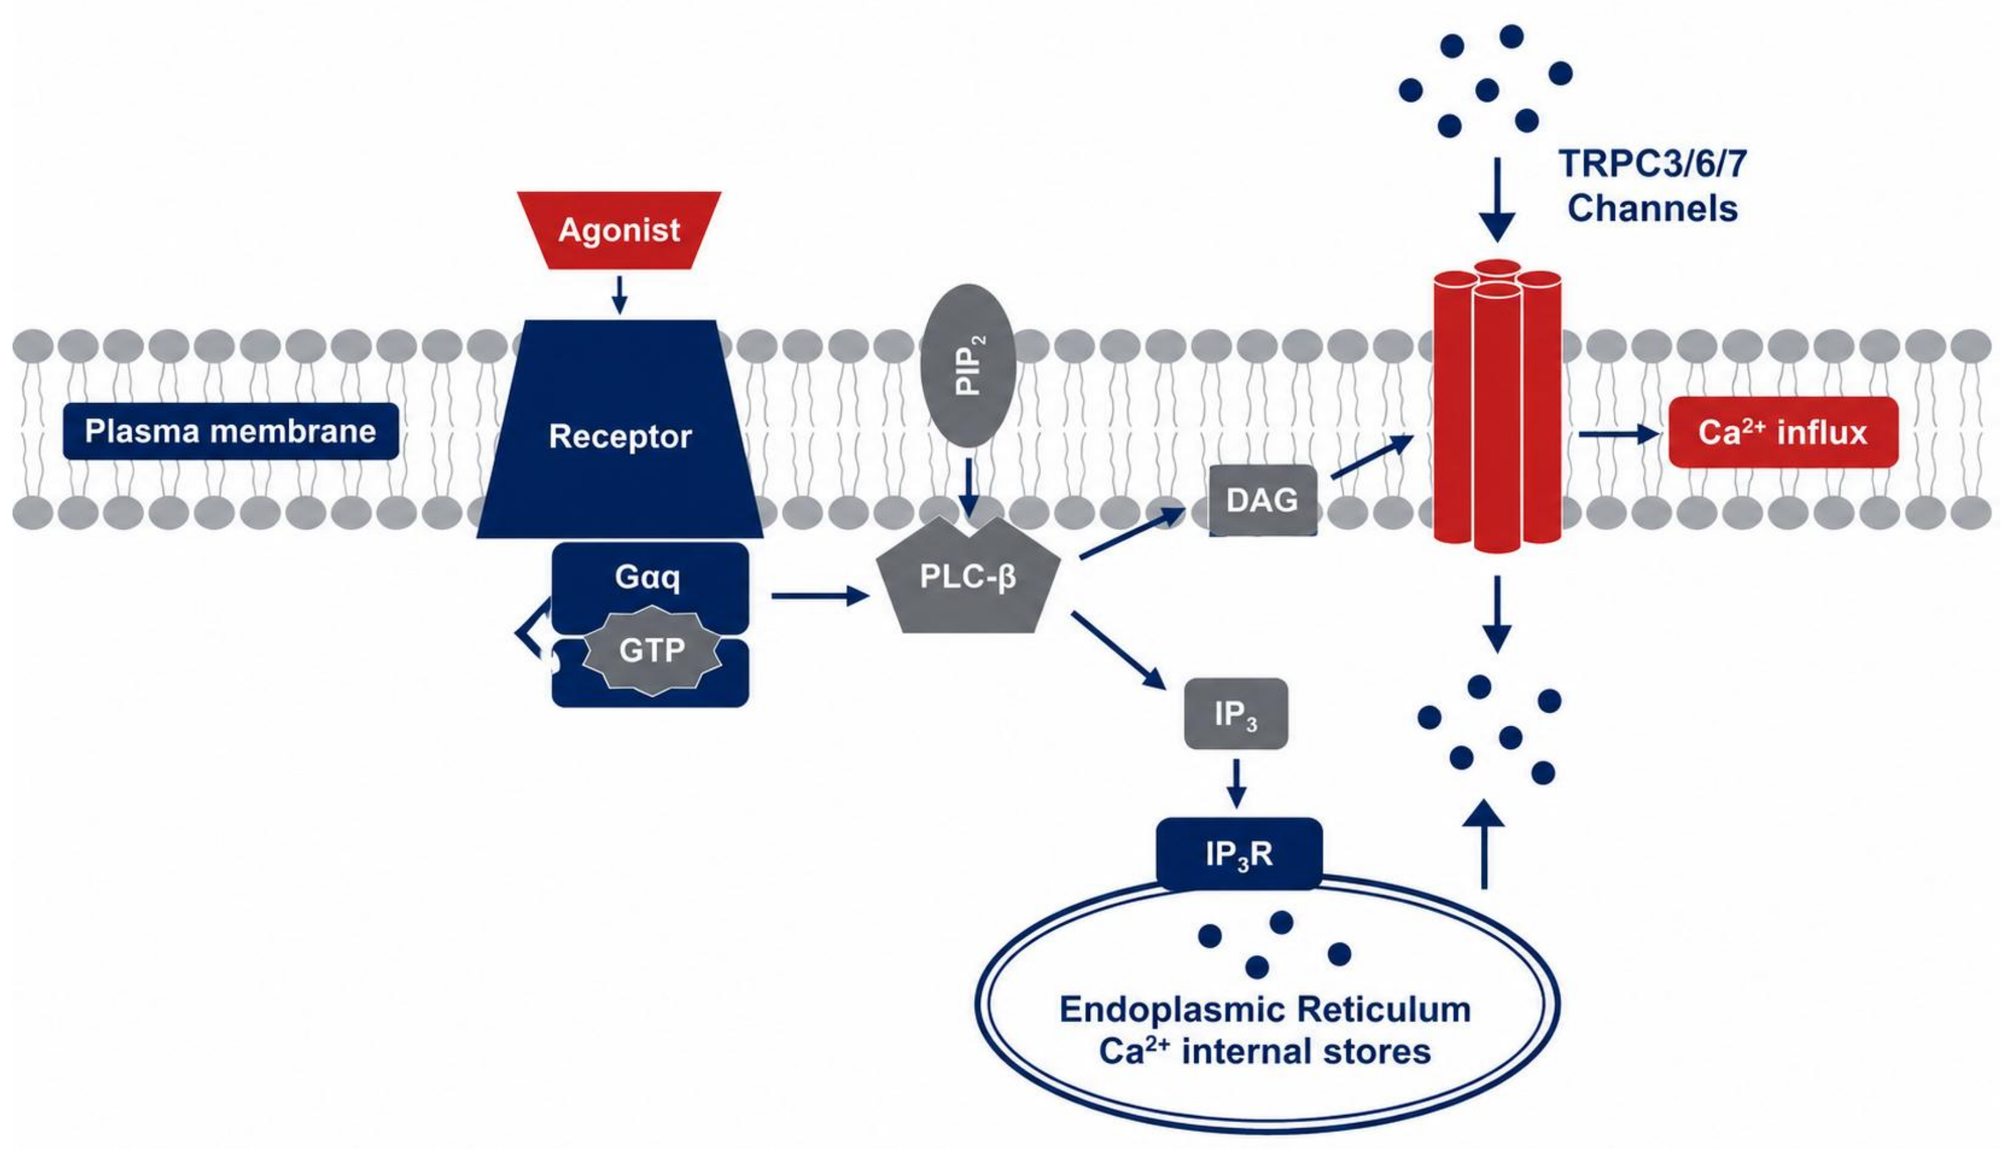
Agonist
Plasma membrane
Receptor
PIP2
Gαq
GTP
PLC-β
DAG
IP3
IP3R
TRPC3/6/7 Channels
Ca2+ influx
Endoplasmic Reticulum
Ca2+ internal stores

Cardiac Cell	TRPC Activation	Potential Xolatrp Effect
Cardiomyocytes	Cell injury	Preserved function
Endothelial cells	Endothelial dysfunction	Improves perfusion
Fibroblasts	Fibrosis	Limits remodelling

Target engagement – Xolatryp inhibits human TRPC channels

Dose-response inhibition of calcium influx in HEK-293 cells

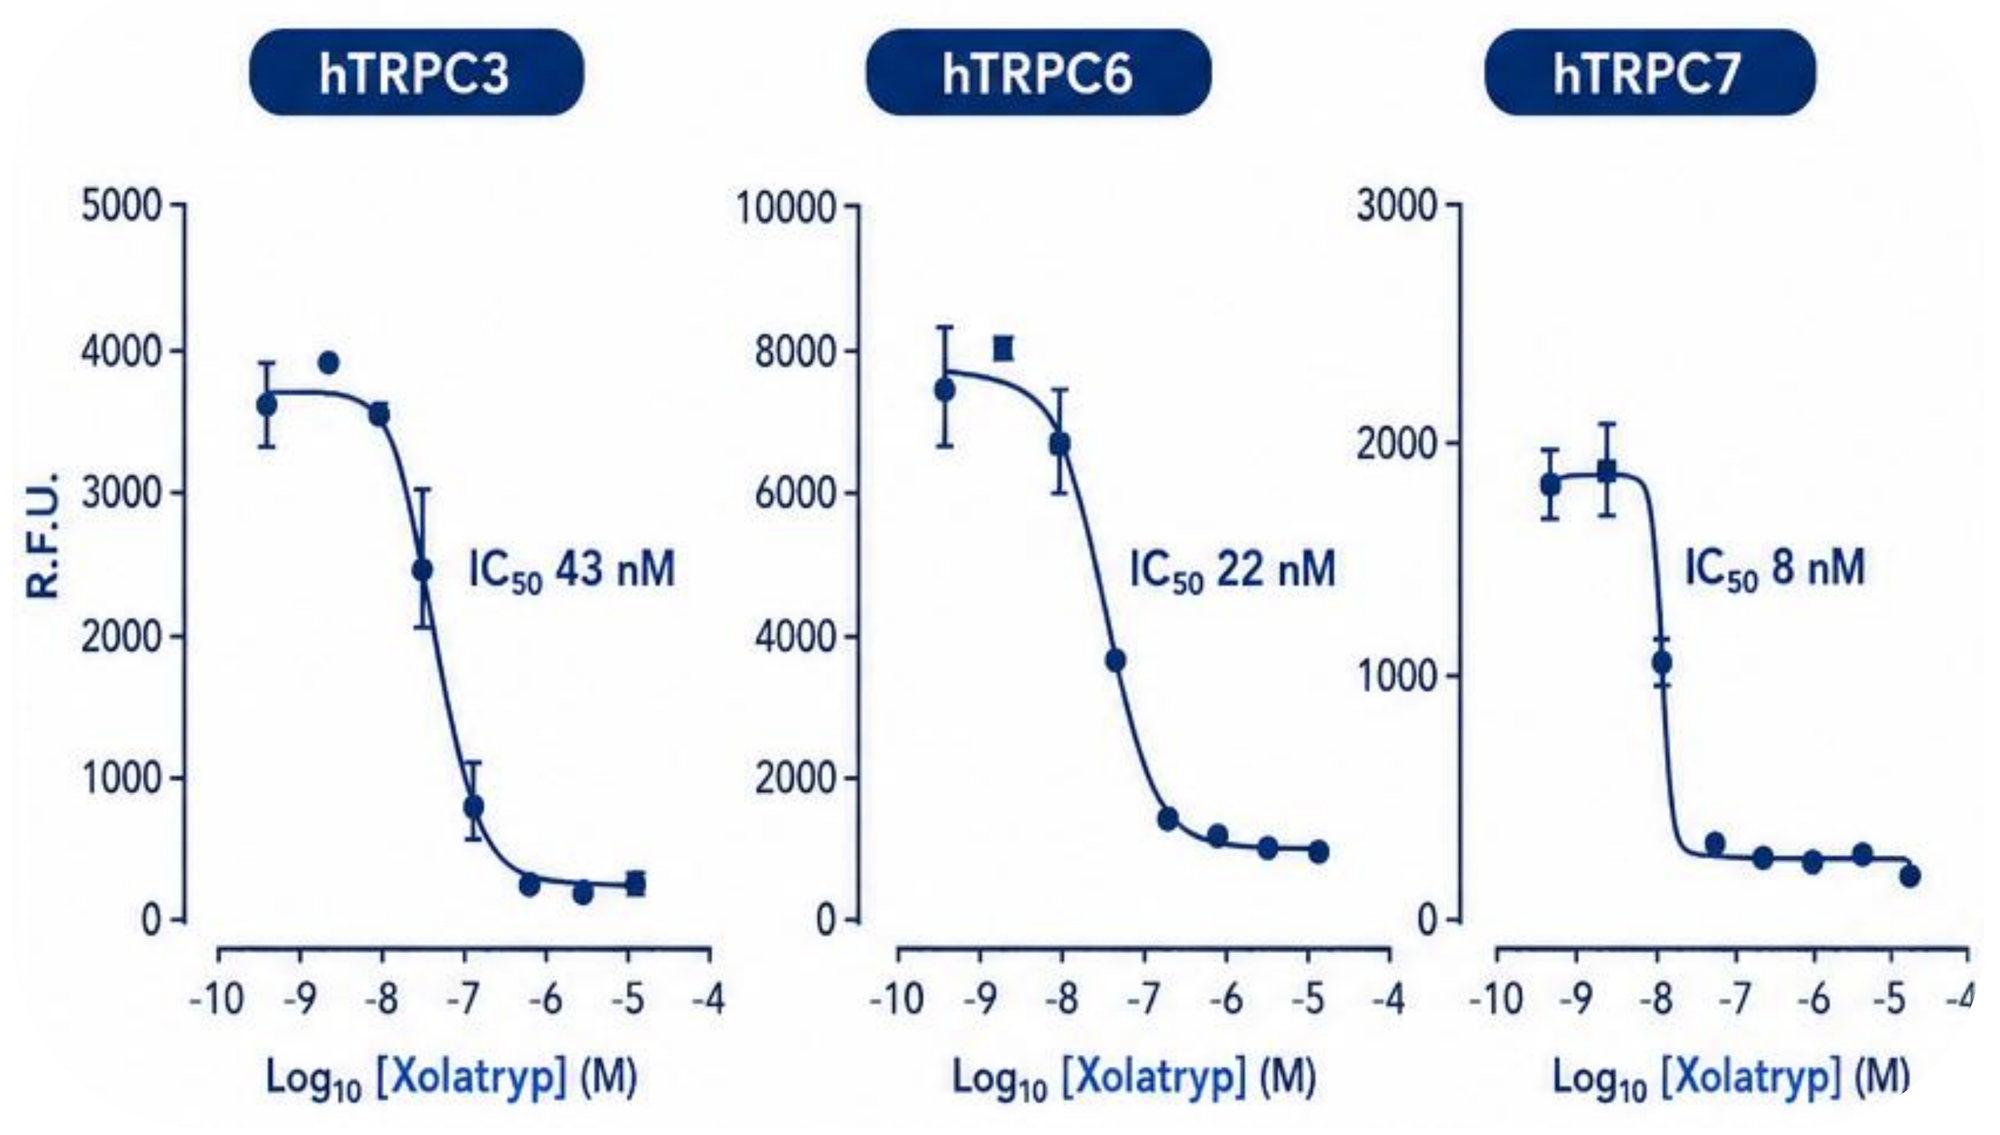
hTRPC3
hTRPC6
hTRPC7
R.F.U.
IC50 43 nM
IC50 22 nM
IC50 8 nM
Log10 [Xolatryp] (M)

Xolatryp inhibits human TRPC3, TRPC6, TRPC7 channels in a concentration-dependent manner

Experimental Design

Generic experimental paradigm for rat myocardial ischaemia-reperfusion studies

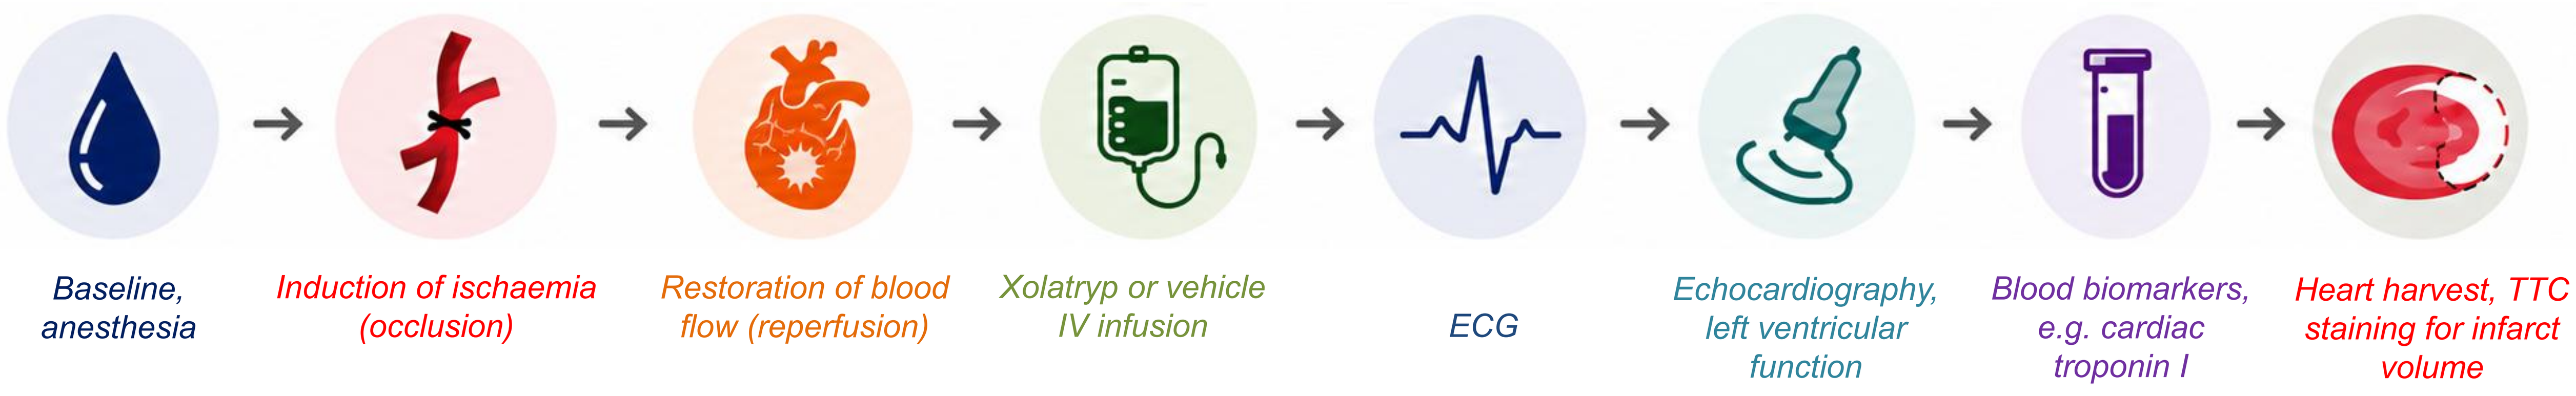
Baseline, anesthesia
Induction of ischaemia (occlusion)
Restoration of blood flow (reperfusion)
Xolatryp or vehicle IV infusion
ECG
Echocardiography, left ventricular function
Blood biomarkers, e.g. cardiac troponin I
Heart harvest, TTC staining for infarct volume

Study A - Proof of Biology

Continuous IV Administration of Xolatryp Preserves Myocardial Structure and Function Following Acute Myocardial Ischaemia

Study Overview - Rat Myocardial Ischaemia-Reperfusion Model

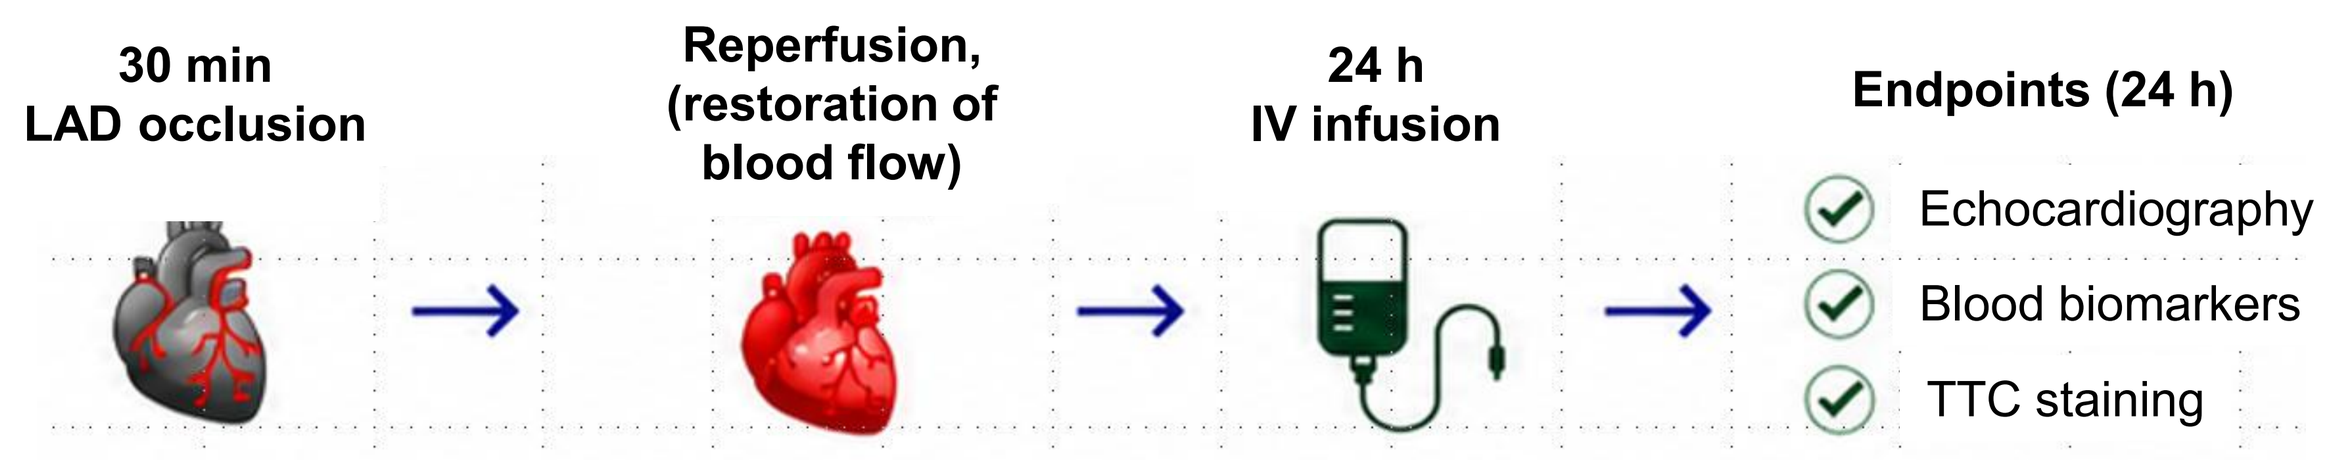
30 min LAD occlusion
Reperfusion, (restoration of blood flow)
24 h IV infusion
Endpoints (24 h)
Echocardiography
Blood biomarkers
TTC staining

Study Details

- Male Sprague-Dawley Rats
- n = 8 per group
- Xolatryp 1.25 mg/kg/h IV infusion
- Infusion commenced 5 min before reperfusion and continued for 24 h

Treatment Groups

Sham, vehicle	I/R, vehicle	I/R, 1.25 mg/kg h Xolatryp (IV)	I/R, 30 mg/kg Captopril (oral)

Preservation of Myocardial Structure by Xolatryp

TTC-stained Heart Sections (Representative)

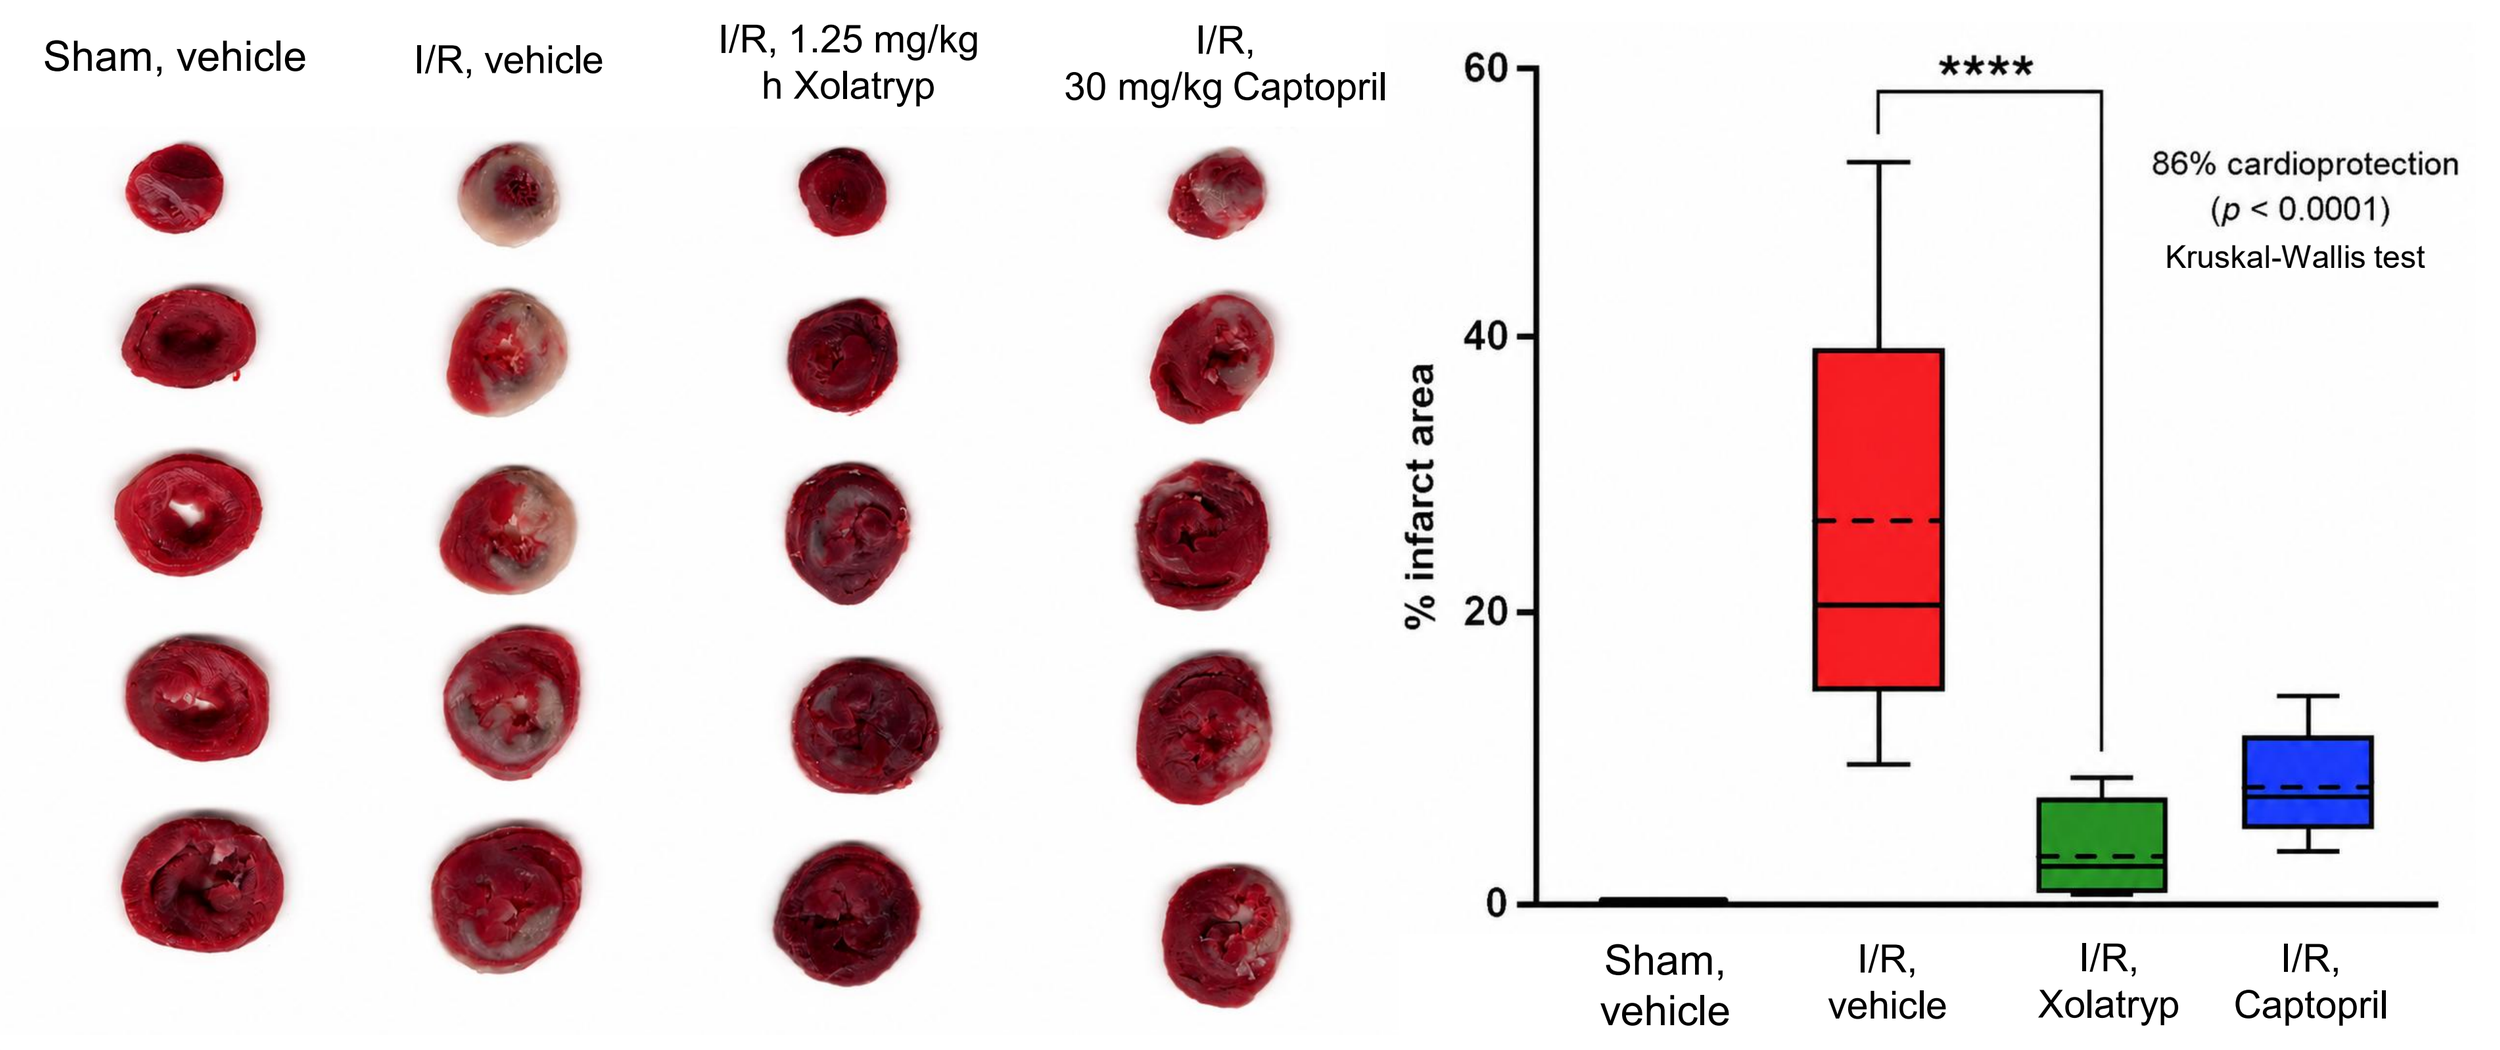
Sham, vehicle
I/R, vehicle
I/R, 1.25 mg/kg h Xolatryp
I/R, 30 mg/kg Captopril
% infarct area

86% cardioprotection (p < 0.0001) Kruskal-Wallis test
Sham, vehicle
I/R, vehicle
I/R, Xolatryp
I/R, Captopril

Preservation of Cardiac Function by Xolatryp (Echocardiography)

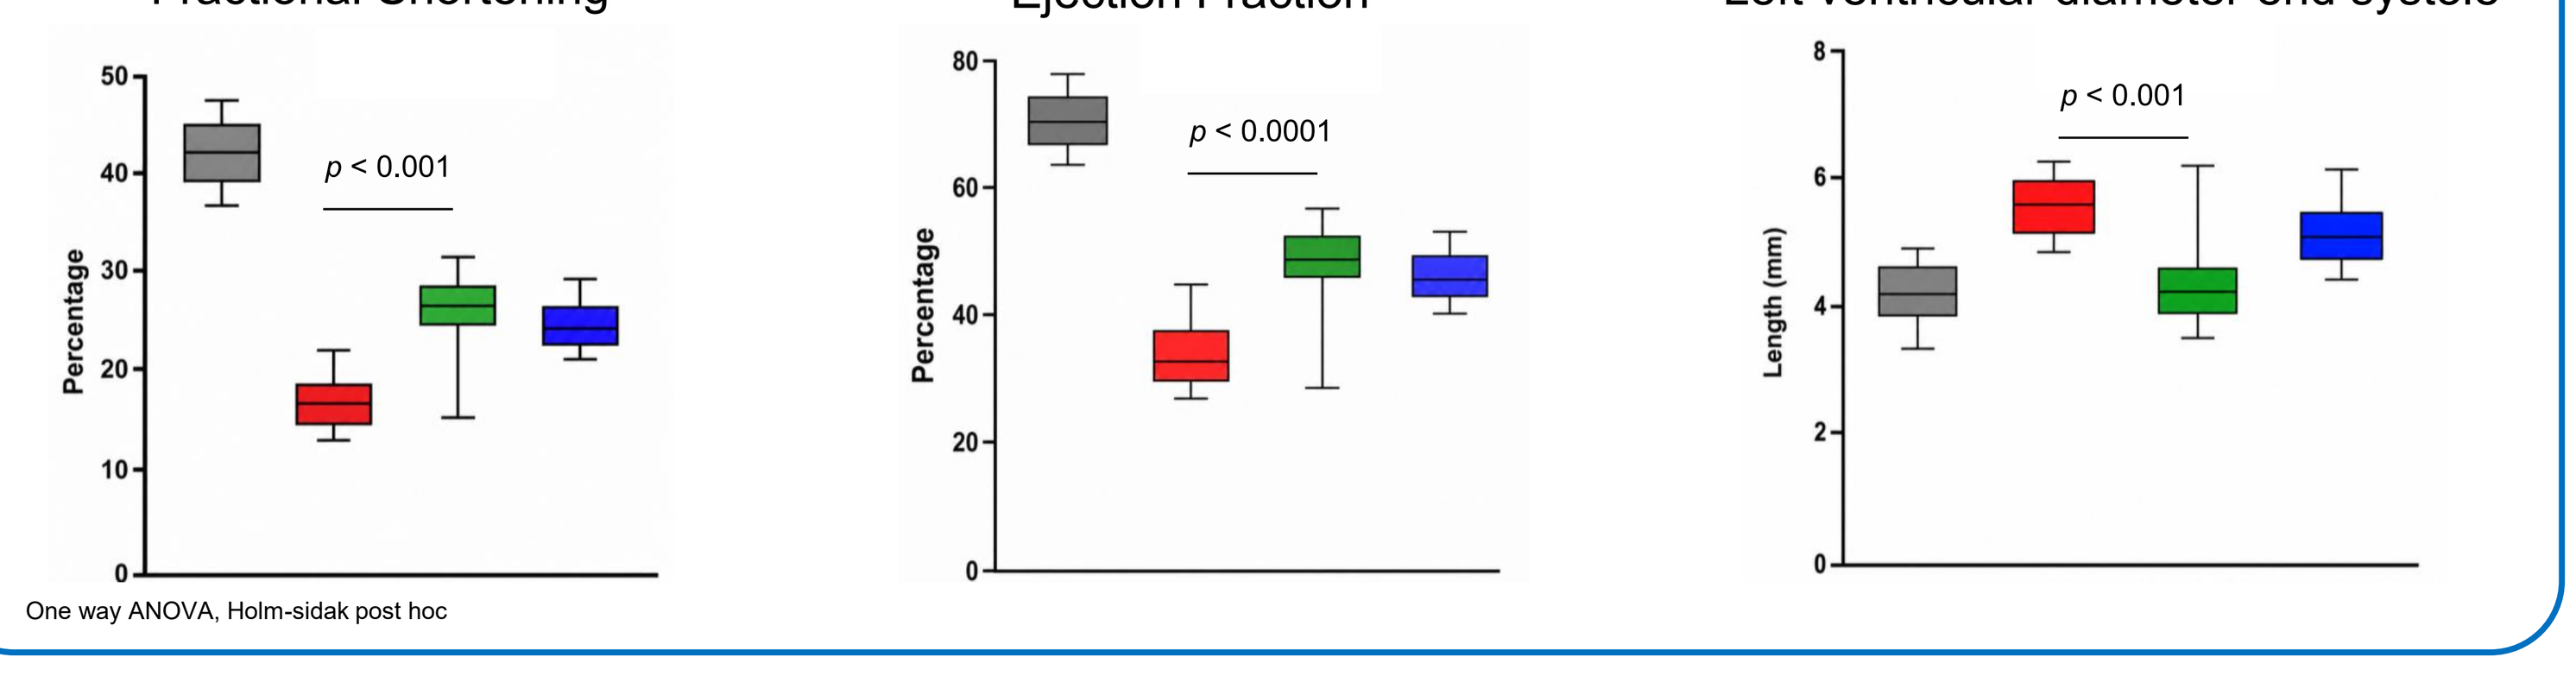
Fractional Shortening
Ejection Fraction
Left ventricular diameter end systole
Percentage
Length (mm)
p < 0.001
p < 0.0001
p < 0.001
One way ANOVA, Holm-sidak post hoc

Plasma Biomarkers of Cardiac and Liver Injury

Xolatryp-Induced Reduction vs I/R Vehicle (%)

Cardiac Troponin I (cTnI)	AST (Liver Biomarker)	Lactate Dehydrogenase
↓ 32% ($p = 0.18$)	↓ 45%** (** $p < 0.05$)	↓ 42%** (** $p < 0.05$)

Key Finding

Continuous IV administration of Xolatryp immediately before reperfusion produced profound cardioprotection, reducing myocardial infarct size by **86%**, preserving left systolic function and attenuated biochemical markers of injury

Abbreviations: LAD, left anterior descending coronary artery; TTC, 2,3,5-triphenyltetrazolium chloride; AST, aspartate aminotransferase; I/R, ischemia/reperfusion; IV, intravenous; cTnI, cardiac troponin I.

Study B - Proof of Translation

Short Duration IV Infusion of Xolatryp Preserves Myocardium and Cardiac Electrical Stability Following Acute Myocardial Ischaemia in Rats

Study Overview

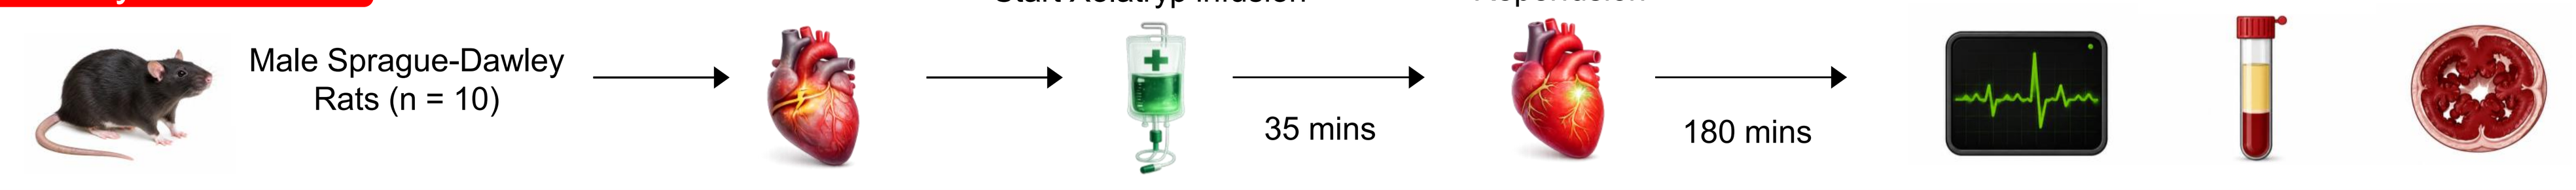
Male Sprague-Dawley Rats (n = 10)
LAD occlusion
Start Xolatryp infusion
35 mins
Reperfusion
180 mins
Assessments

Preservation of Myocardial Tissue (TTC Staining)

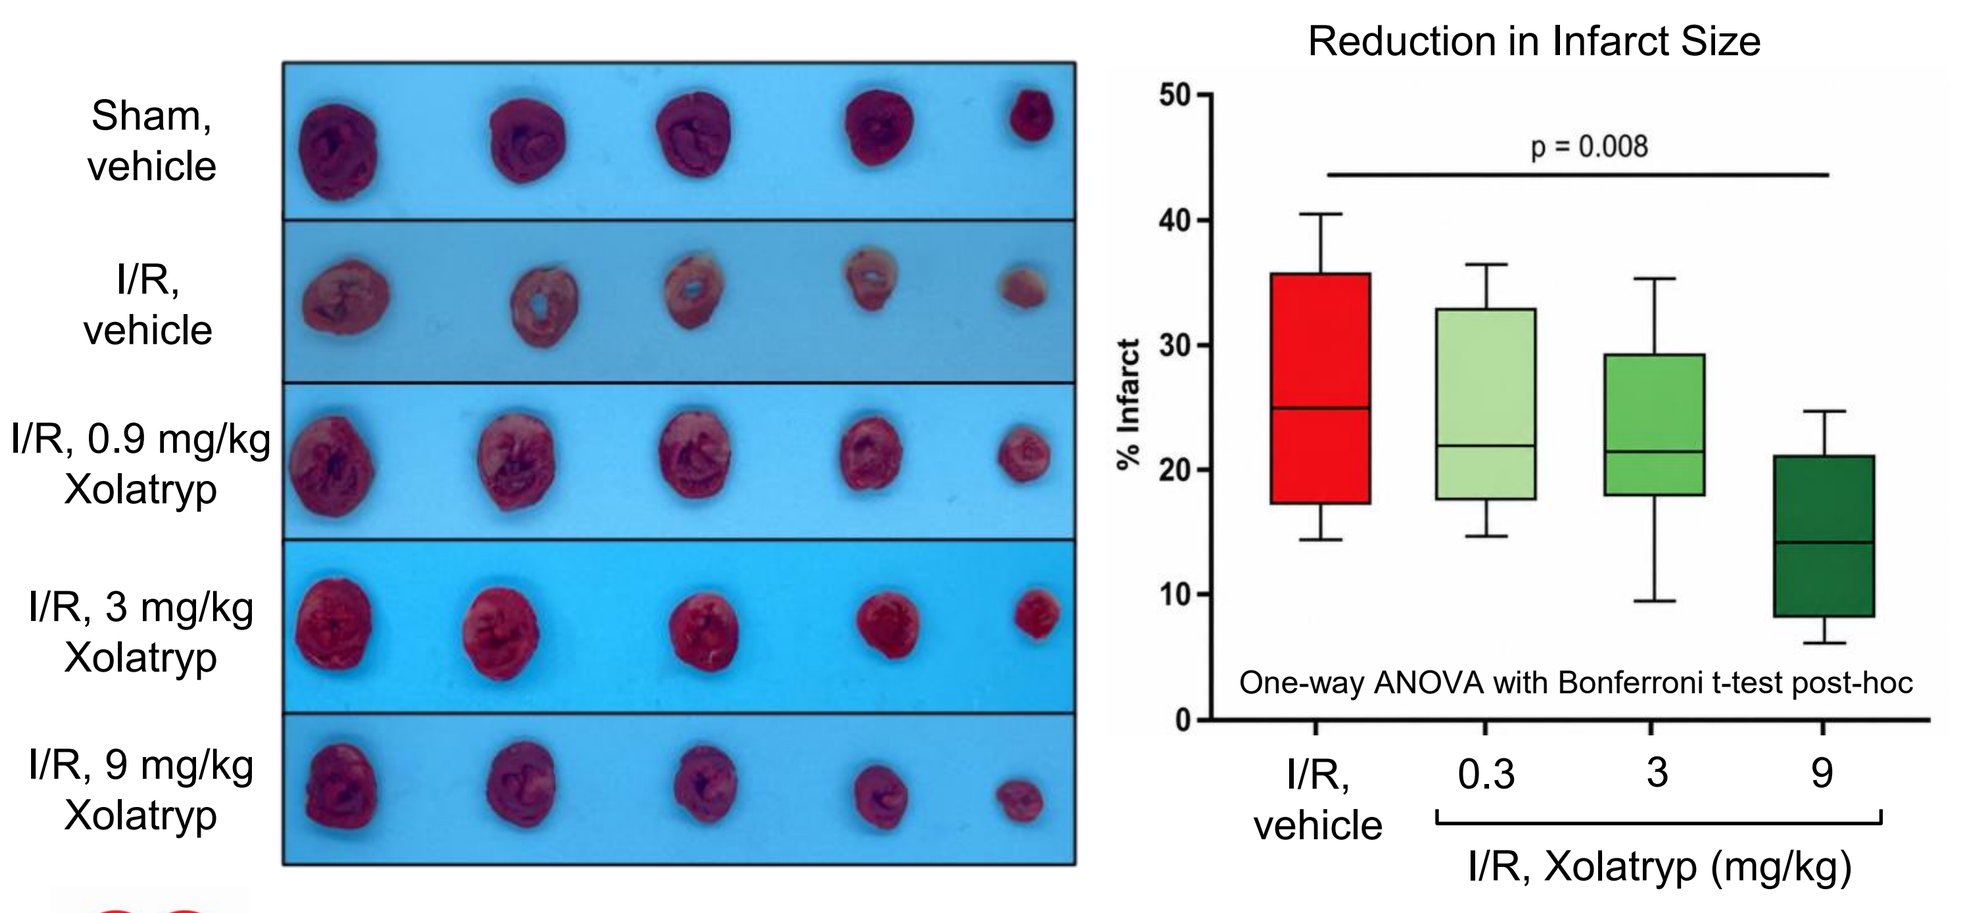
Sham, vehicle
I/R, vehicle
I/R, 0.9 mg/kg Xolatryp
I/R, 3 mg/kg Xolatryp
I/R, 9 mg/kg Xolatryp
Reduction in Infarct Size
p = 0.008
% Infarct
One-way ANOVA with Bonferroni t-test post-hoc
I/R, vehicle
0.3
3
9
I/R, Xolatryp (mg/kg)

42% reduction in infarct size at 9 mg/kg Xolatryp versus I/R vehicle

Reduced Plasma Biomarkers

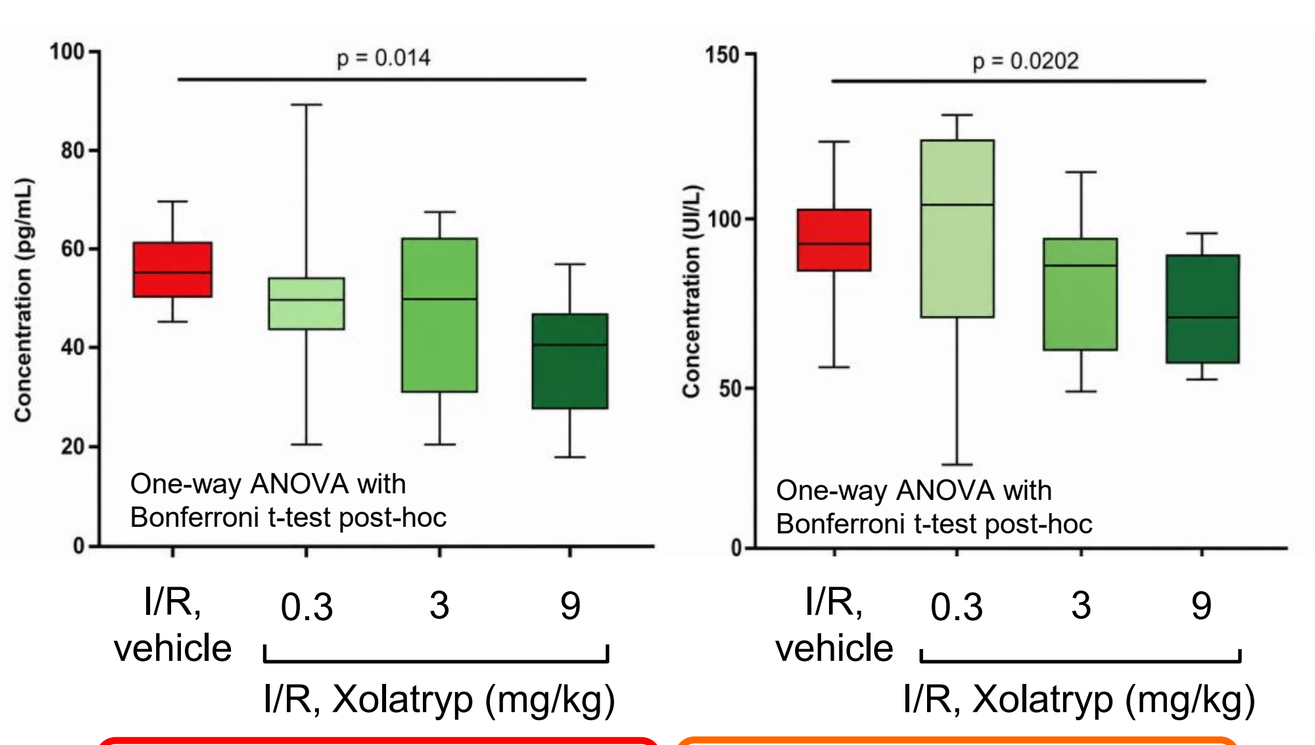
Cardiac Troponin I (cTnI)
Alanine Transferase (ALT)
p = 0.014
p = 0.0202
Concentration (pg/mL)
Concentration (UI/L)
One-way ANOVA with Bonferroni t-test post-hoc
I/R, vehicle
0.3
3
9
I/R, Xolatryp (mg/kg)

Cardiac Troponin I	Alanine Transferase
↓ 32% ($p = 0.014$)	↓ 21% ($p = 0.0202$)

9 mg/kg Xolatryp-Induced Reduction vs I/R Vehicle (%)

Reduced Ventricular Premature Beats (VPBs)

Continuous ECG Monitoring During 3 h Reperfusion After Ischaemia

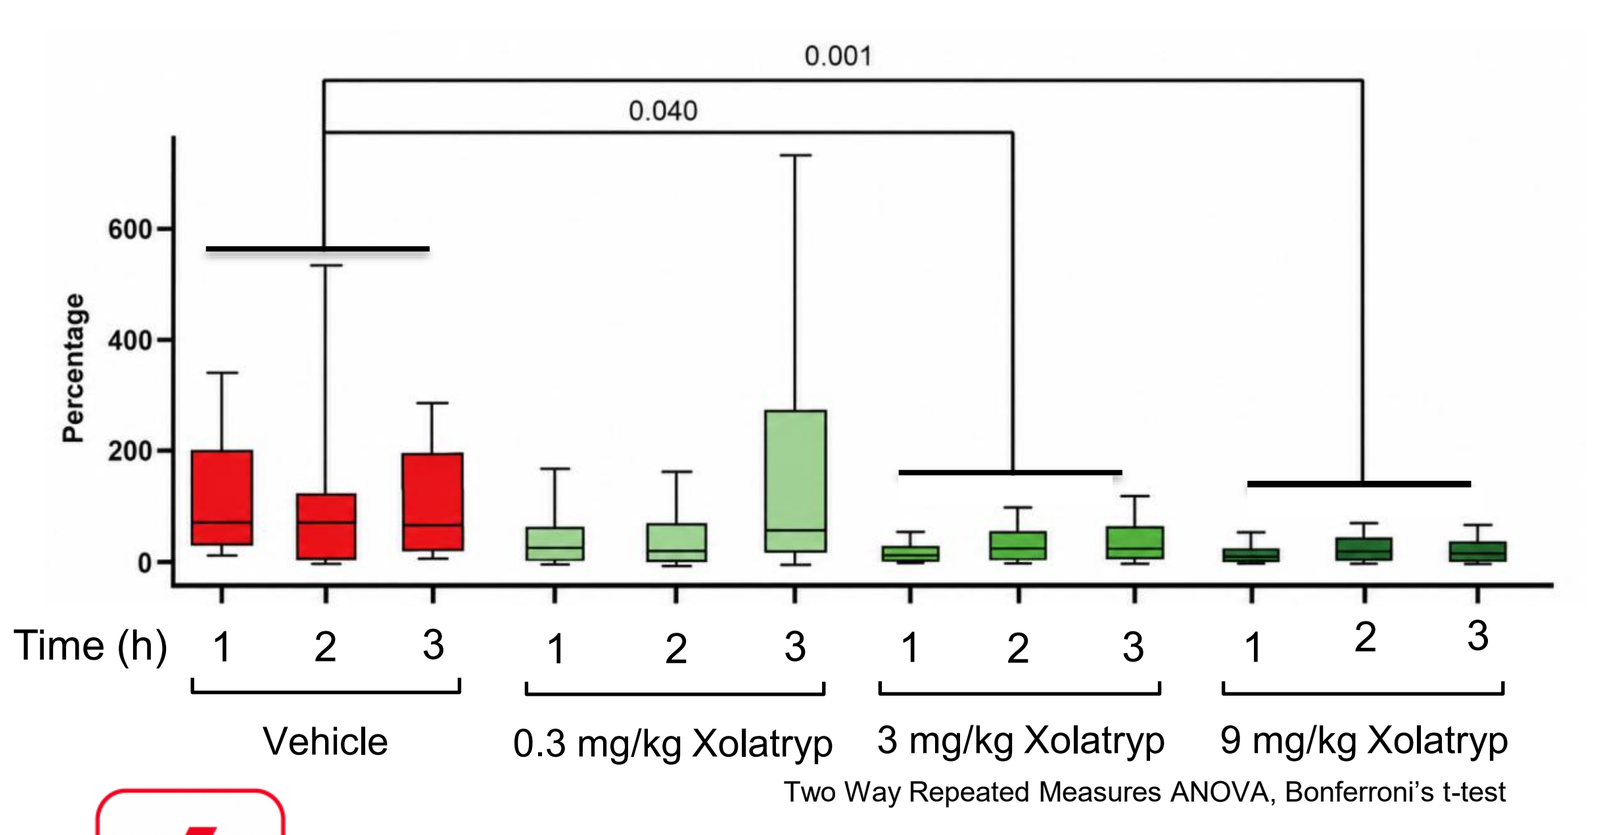
0.001
0.040
Percentage
Time (h)
Vehicle
0.3 mg/kg Xolatryp
3 mg/kg Xolatryp
9 mg/kg Xolatryp
Two Way Repeated Measures ANOVA, Bonferroni's t-test

90% reduction in VPBs by 9 mg/kg Xolatryp versus I/R vehicle at 3 h reperfusion

Decreased Ventricular Tachycardia and Fibrillation (VT/VF)

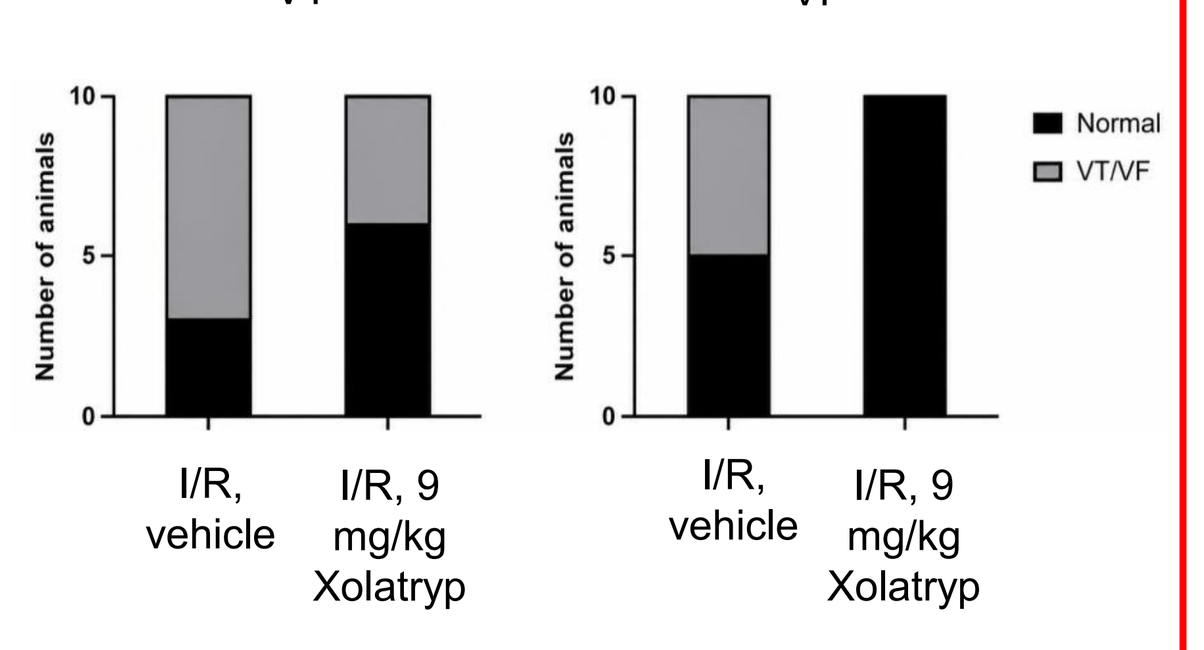
VT
VF
Number of animals
Normal
VT/VF
I/R, vehicle
I/R, 9 mg/kg Xolatryp

Xolatryp completely suppressed VFs and markedly reduced VTs throughout 3 h of reperfusion

Abbreviations: LAD, left anterior descending coronary artery; TTC, 2,3,5-triphenyltetrazolium chloride; AST, aspartate aminotransferase; I/R, ischemia/reperfusion; IV, intravenous; cTnI, cardiac troponin I.

Key Finding

A short IV administration of Xolatryp dose-dependently produced cardioprotection, reduced cardiomyocyte injury, suppressed ventricular ectopy and curtailed malignant ventricular arrythmias after myocardial ischaemia-reperfusion injury

Clinical Translation

➢ The safety and tolerability of Xolatryp was evaluated in a Phase I First-in-Human (FIH) study (NYR-BI03-01) as an IV infusion for up to 6 h. Xolatryp demonstrating a favourable safety profile
➢ The compelling cardioprotective and anti-arrhythmic efficacy observed in complementary rat myocardial ischaemia-reperfusion models directly informed the design of the ongoing PROTECT-MI Phase IIa clinical trial (https://clinicaltrials.gov/study/NCT07362446?term=PROTECT-MI&viewType=Card&rank=4).
➢ PROTECT-MI is a first-in-patient, randomised, double-blind, placebo-controlled, multicentre study evaluating whether a single 6-hour intravenous infusion of Xolatryp, administered as an adjunct standard of care in STEMI patient undergoing primary PCI is safe and well tolerated. Efficacy is explored as secondary and exploratory end points.
➢ Clinical outcomes will measure the incidence of AEs and SAEs with efficacy redouts including infarct size, ejection fraction, fractional shortening, serial blood cardiac biomarkers, arrhythmia monitoring and safety assessments to determine whether the cardioprotective effects observed preclinically translate to patients undergoing reperfusion therapy.

Acknowledgements
The authors gratefully acknowledge the contributions of:
- Pharmaron, Inc. (Louisville, Kentucky) for conducting Study A.
- Sichuan Greentech Biotechnology Co., Ltd. (Chengdu, China) for conducting Study B.
- Sygnature Discovery Ltd. (Nottingham, United Kingdom) for undertaking the in vitro TRPC3/6/7 channel pharmacology and concentration–response assays.
- The investigators, research staff and patients participating in the ongoing PROTECT-MI Phase IIa clinical trial.

Disclosures
This work was funded by Nyrada Inc. B.E., J.P., A.S., P.C. and J.B. are employees and/or shareholders of Nyrada Inc. G.D. Housley serves as a scientific advisor to Nyrada Inc. and shareholder. Xolatryp® is an investigational medicinal product being developed by Nyrada Inc.

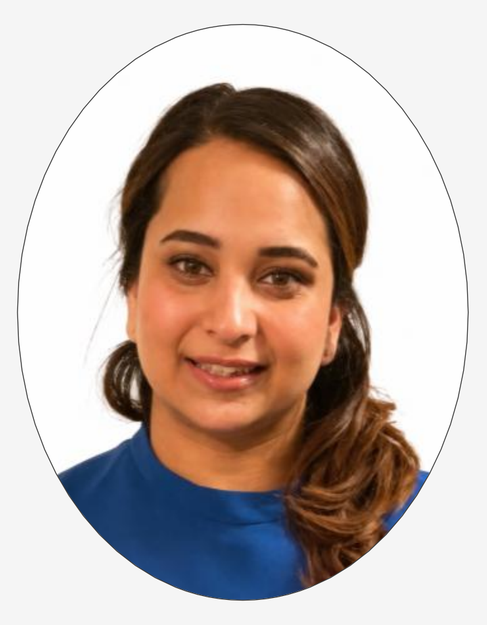

Presenter:
Dr Jasneet Parmar
Director, R&D
Nyrada Inc